How does the body heal itself?

Our bodies are subjected to constant wear and tear, injury and damage.

What happens when we get injured, or when things just wear out?

If you find your car window broken 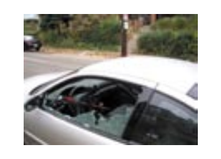 you can fix it by grabbing the duct tape or getting a new window 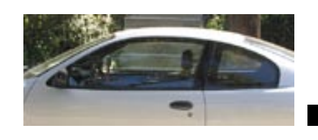.

After injury to a tissue in the body, cells from the immune system 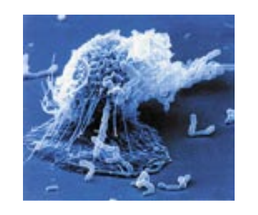 clean up, and the tissue heals by a combination of two ways

Repair is when the body grabs its duct tape, the fibroblast 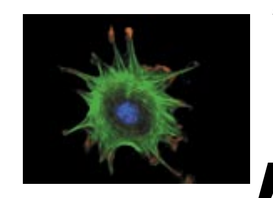, and fills in the damaged tissue with something tough and strong, but otherwise incapable of doing what the damaged tissue once did, a scar.

Regeneration is the complete restoration of the original tissue. At the end of the healing process, the damaged tissue is replaced with new functional cells. Like a replaced window, it can be difficult to tell what happened.

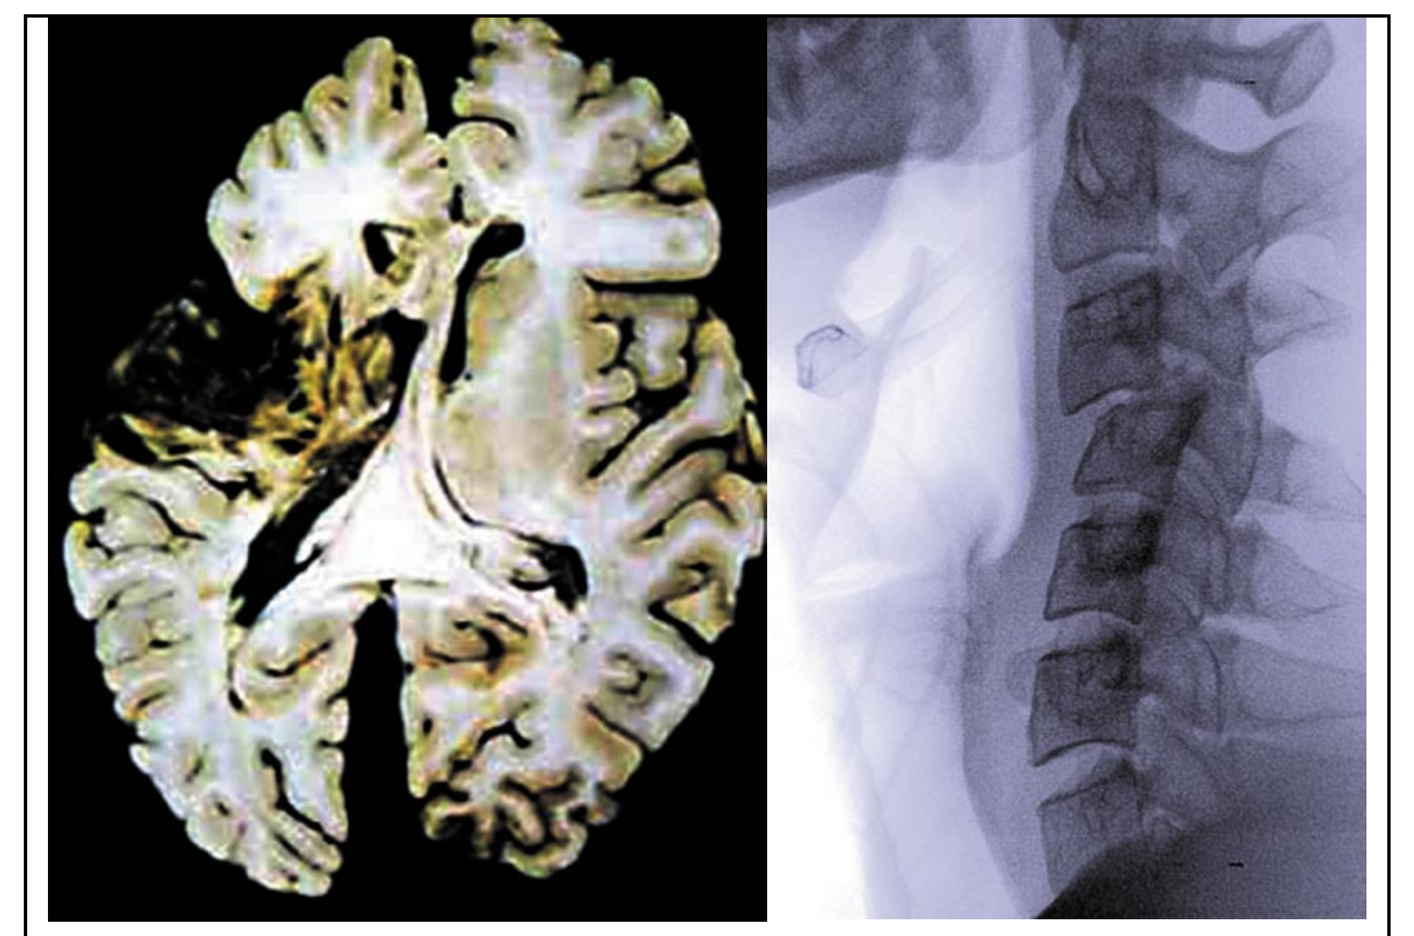

A stroke or spinal cord injury heals almost entirely by repair. The multitude of connections destroyed cannot easily be recreated, nor are the cells replaced.

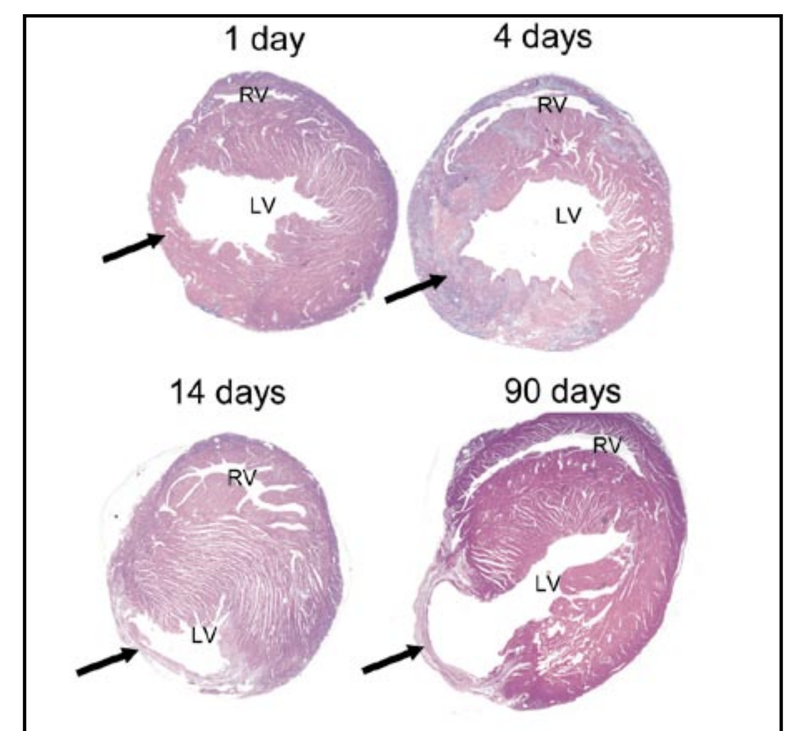
1 day
4 days
14 days
90 days
RV
LV

After a heart attack what was once a thick muscular wall is gradually replaced by a thin immobile scar.

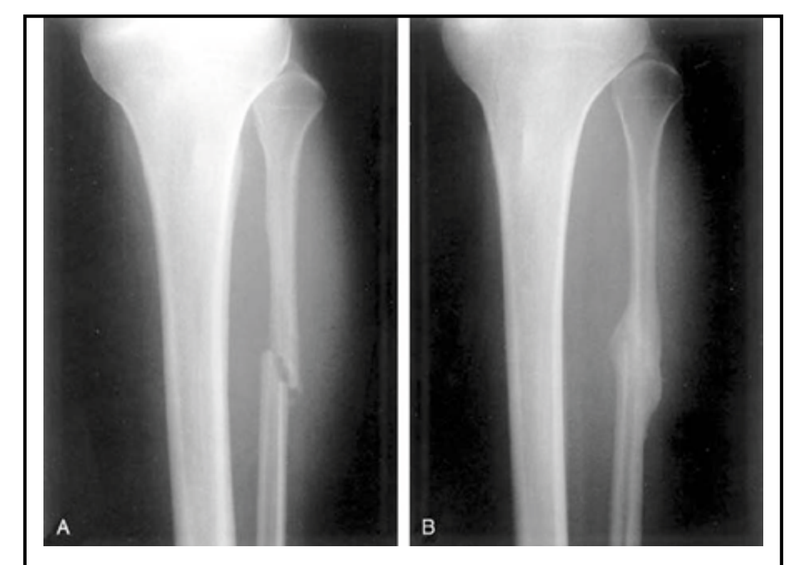
A
B

A broken bone often heals by regenerating something close to the original bone. All that the doctor has to do is get the remaining fragments of bone aligned close enough to the original bone.

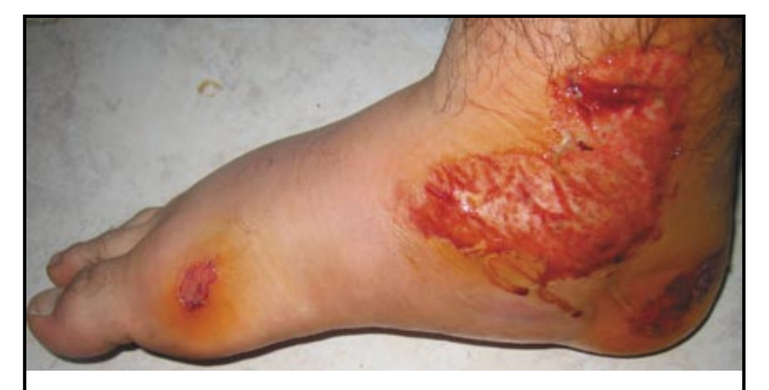

Our skin can heal mostly by regeneration. After an abrasion, one can watch the new skin grow in from both the edges of the wound and from islands of new skin growing up from where the hair once was.

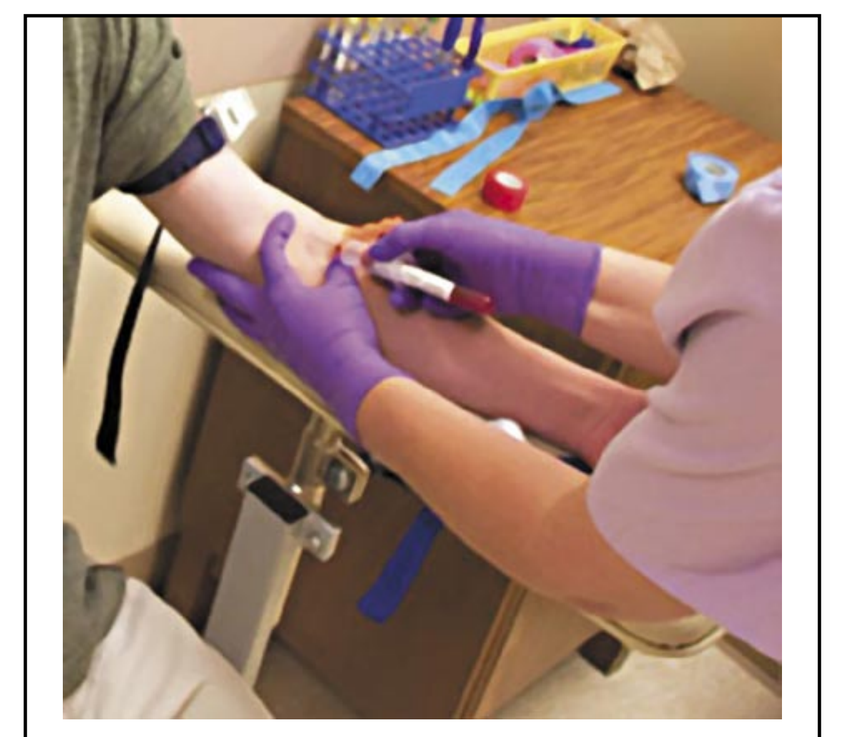

Blood perfectly regenerates after loss. Within only a few days or weeks it is impossible to tell any blood was lost.

Injured tissues **heal by a blend of repair and regeneration.** In the more regenerative tissues after a while it's hard to tell injury occurred at all. In tissues where regeneration doesn't occur, or is weak, the injured tissue either disappears or is replaced by scar and no longer works properly.

Why can only some tissues regenerate?

A tissue can **regenerate by** either **dividing the existing functional cells** or by activating **a resident stem cell population** to create new functional cells. If the cells cannot divide, or there are no (or not enough) resident stem cells, the tissue will fail to regenerate, and default to repair. Some tissues, like skin, liver, bones, gut and the blood are constantly turning over. These tissues tend to regenerate the best.

What is a stem cell? A stem cell must be able to make more copies of itself (self-renewal) and give rise to more capable offspring (differentiate).

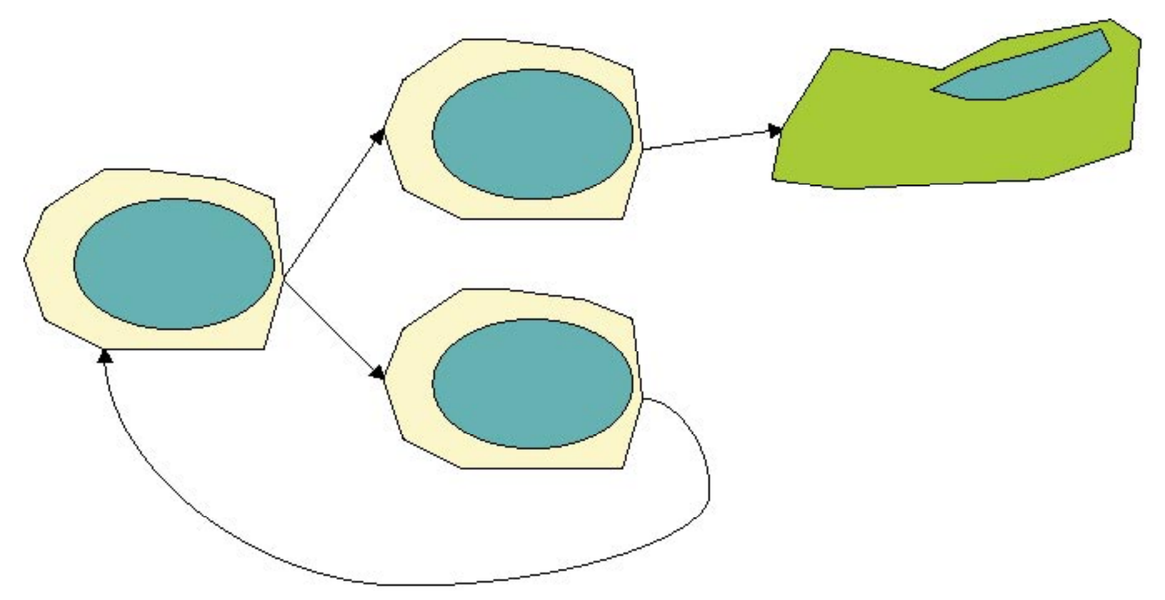

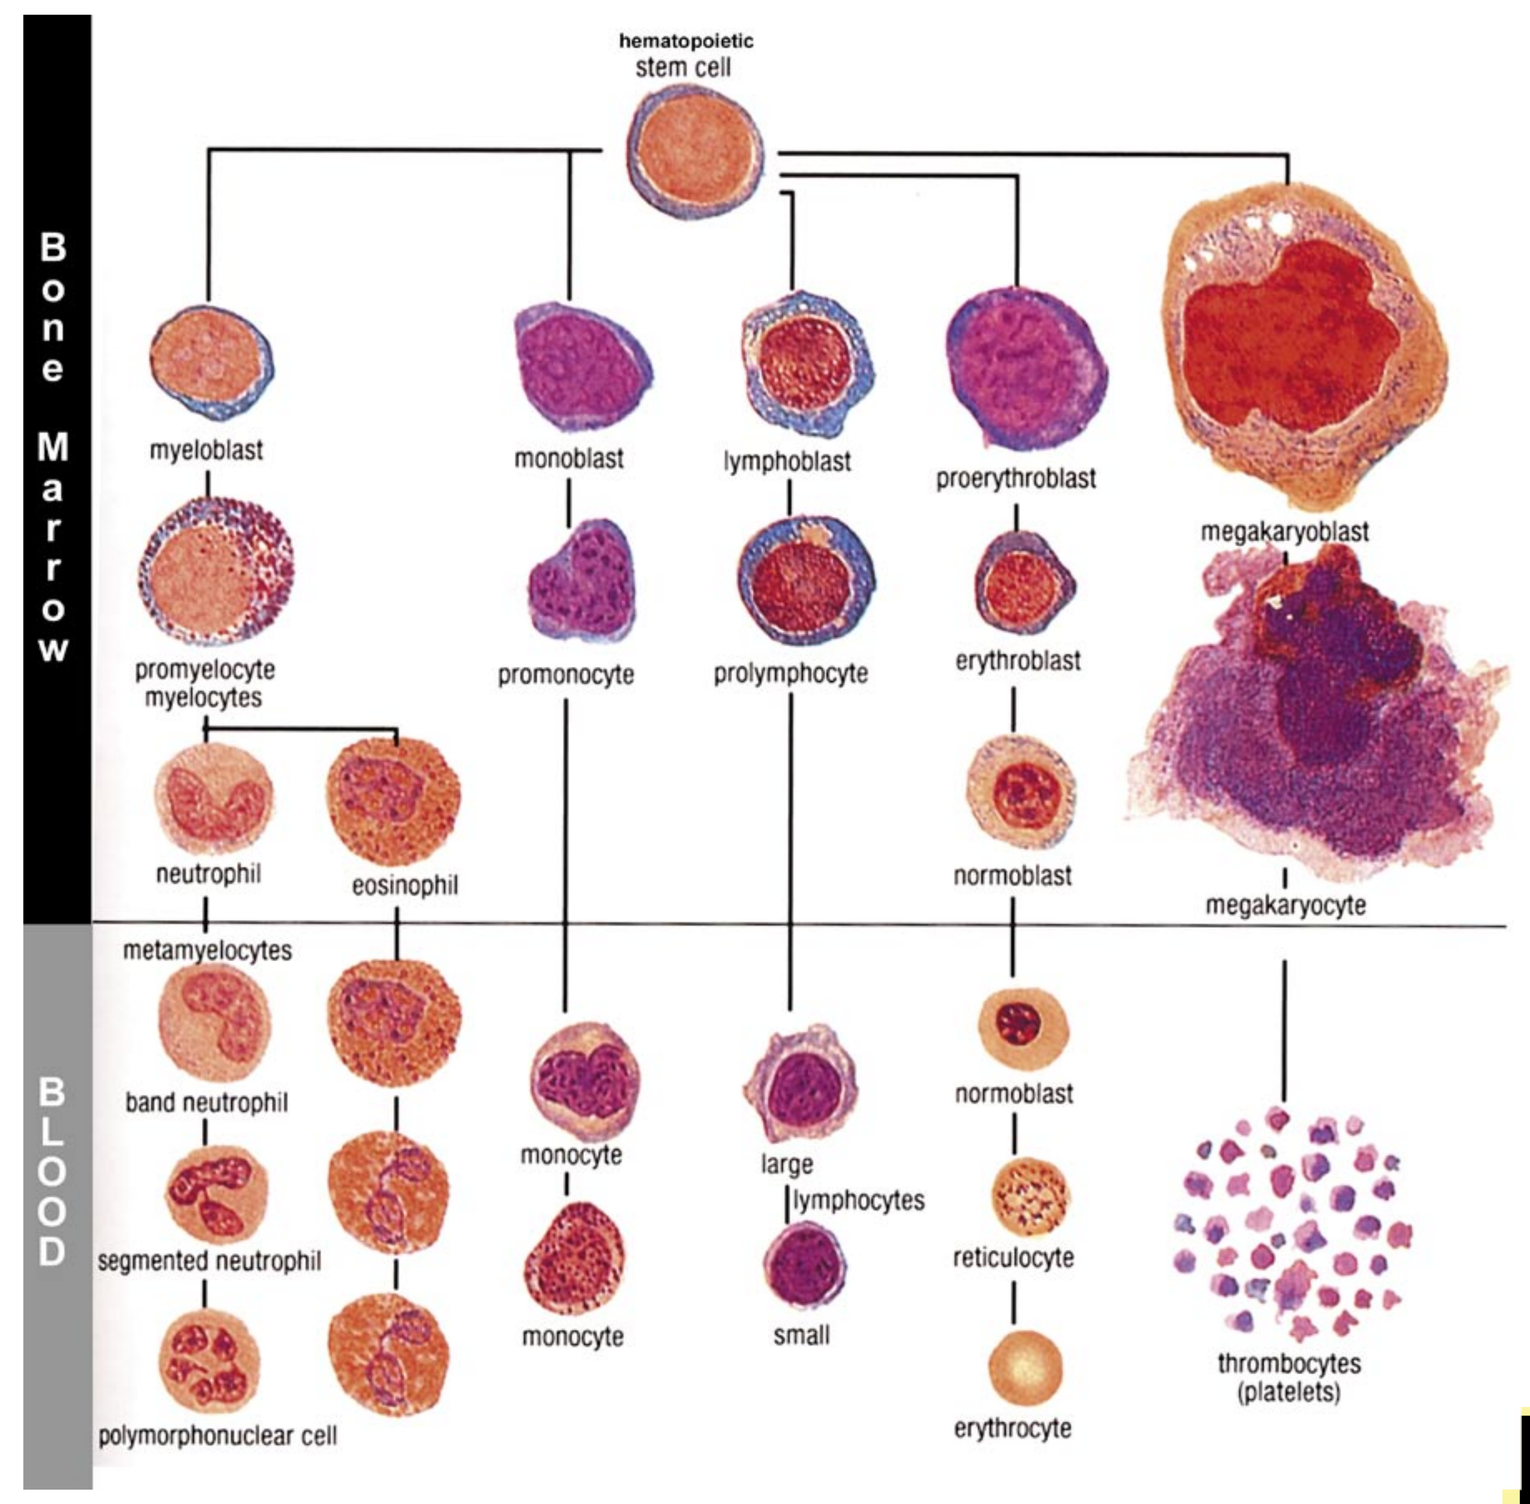
Bone Marrow
BLOOD
hematopoietic stem cell
myeloblast
monoblast
lymphoblast
proerythroblast
megakaryoblast
promyelocyte myelocytes
promonocyte
prolymphocyte
erythroblast
neutrophil
eosinophil
normoblast
megakaryocyte
metamyelocytes
band neutrophil
monocyte
large lymphocytes
normoblast
segmented neutrophil
reticulocyte
thrombocytes (platelets)
polymorphonuclear cell
monocyte
small
erythrocyte

Adult stem cells, like those of the blood, can give rise to all the cells in the tissue from which they came. The more things the cell can do, the fewer different types of cells it can become.

The **embryonic stem cell** is the stem cell of stem cells, capable of becoming any tissue type in the body, including those that lack adult stem cells and cannot regenerate, like heart, brain and spinal cord. 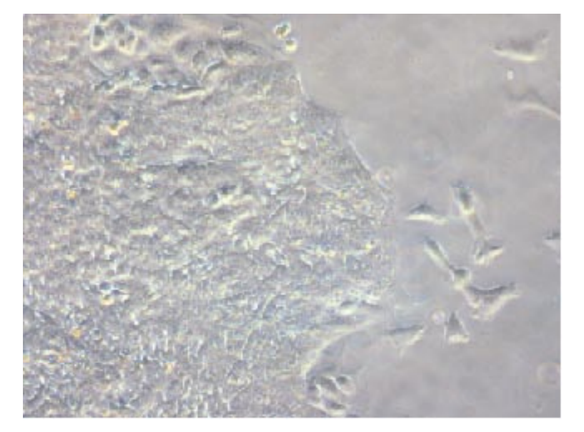

How can we use stem cells to help?

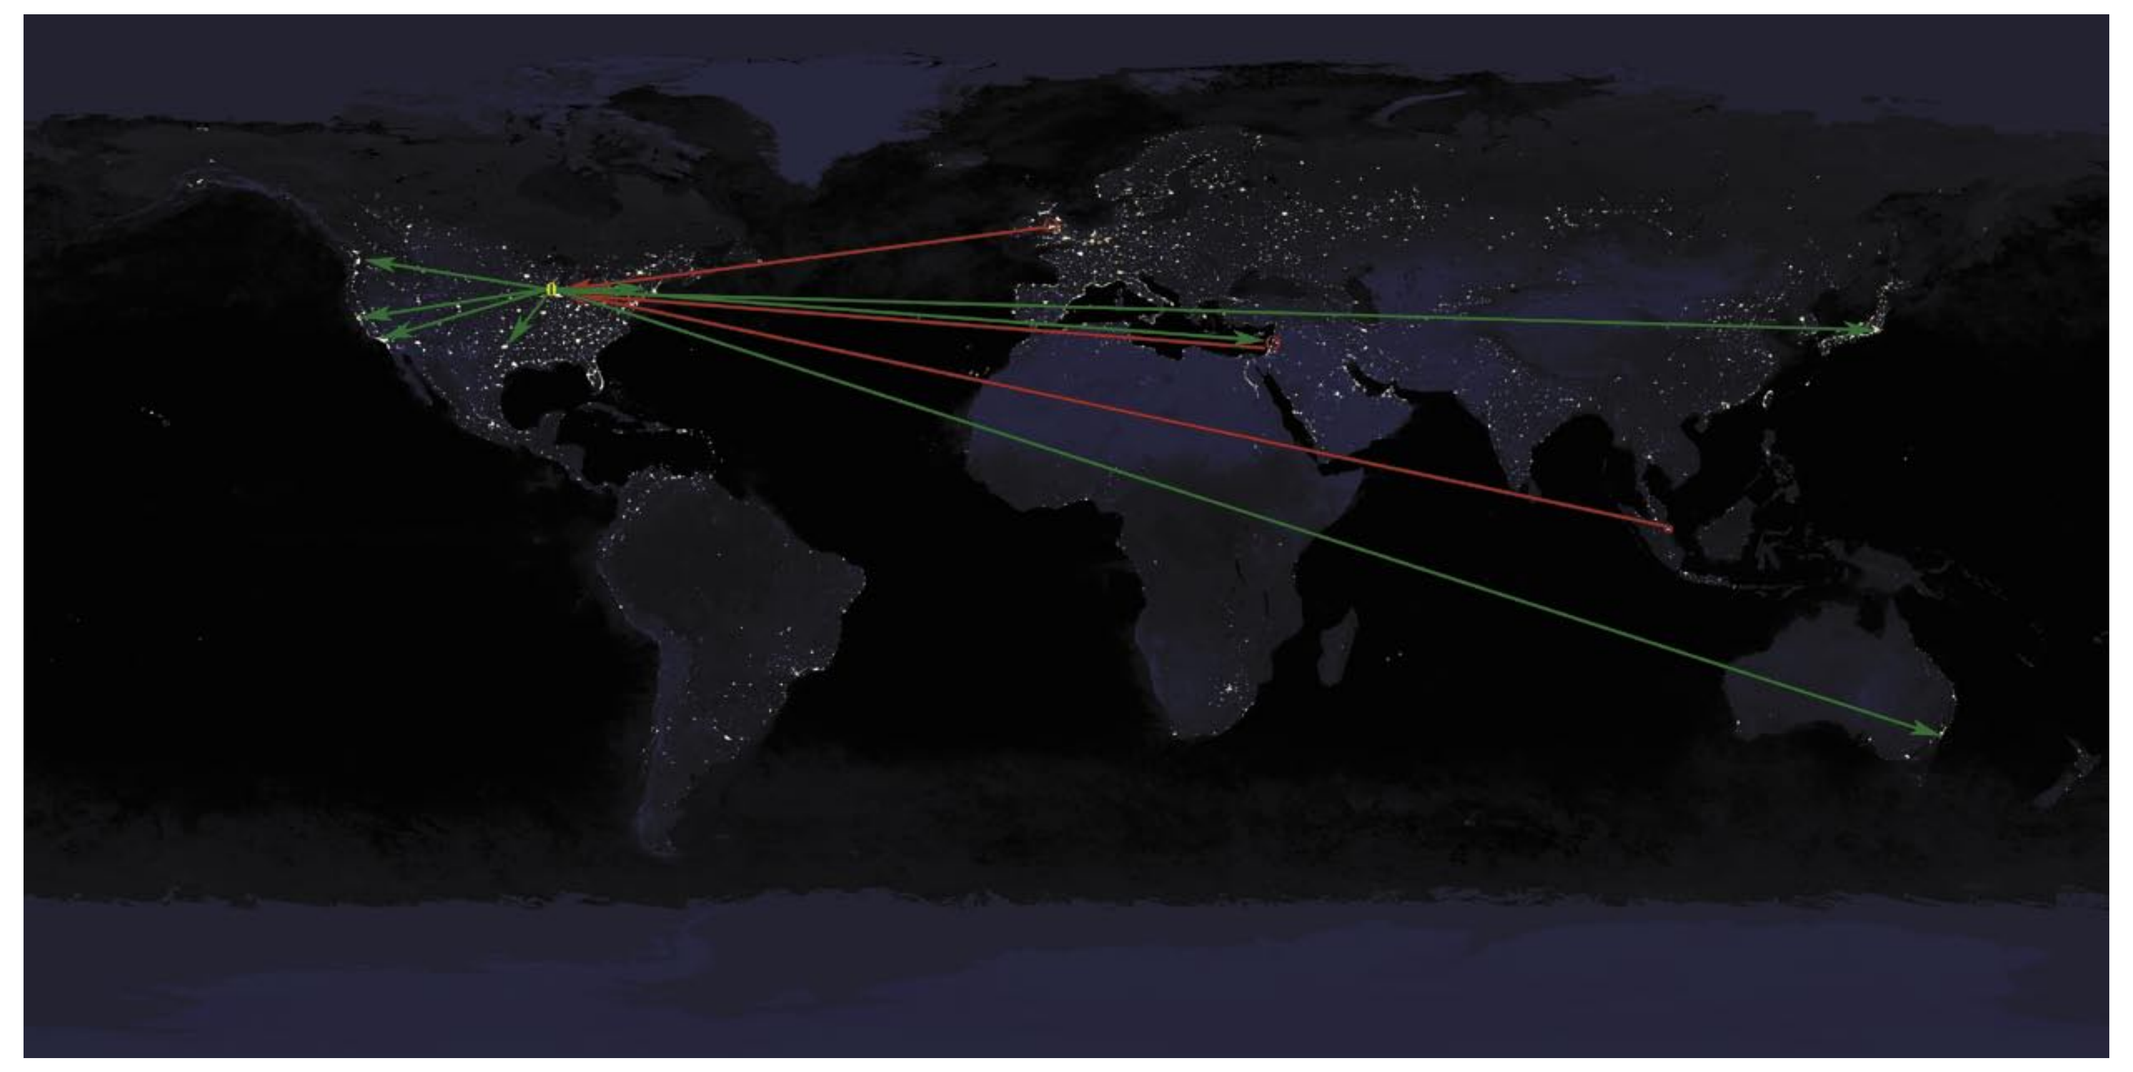

An **international collaboration** helped create the first human embryonic stem cell at the University of Wisconsin, using techniques and reagents from Singapore, Israel and the UK (among others). Since then, the cells and techniques have circled the globe.

The majority of illness and death is caused by damage to tissues that cannot regenerate. By using **embryonic stem cells as a source of replacement cells** scientists hope to help tissues that would otherwise heal by repair to regenerate, such as heart and brain.

Center for Cardiovascular Biology and Regenerative Medicine

Department of Pathology, School of Medicine, University of Washington
Jonathan L. Golob, Amanda M. Masino, Charles Murry.